Aboyans V [1], Miadi H [2], Morboeuf O [2], Piettre E [2], Jolivel R [3], Bura-Rivière A [4]

[1] Department of Cardiology, Dupuytren University Hospital, and INSERM 1094 & IRD, University of Limoges, Limoges, France,
[2] Bayer Healthcare SAS, Loos, France,
[3] Heva, Lyon, France,
[4] Rangueil University Hospital, Toulouse, France

Epidemiology of Patients Revascularized for Peripheral Artery Disease in France (2016-19):

A Retrospective Observational Cohort Study Using the French Hospital Discharge Database (PMSI)

Objectives

There is a lack of up-to-date epidemiological data regarding patients with lower extremity peripheral artery disease (PAD) who underwent lower-limb revascularization in France [1]. The objectives of the study were to quantify the number of patients with PAD who annually undergo lower-limb revascularization in France, and to assess the onset of atherothrombotic events (AE) in this population.

Methods

Using the French Hospital Discharge Database (PMSI), patients hospitalized for revascularization for PAD (index date) were followed from January 1, 2016, to December 31, 2019. AE (a composite of acute limb ischemia, major amputation for vascular cause, myocardial infarction, ischemic stroke/TIA -Transient Ischemic Attack- and in-hospital death) as well as comorbidities were identified using ICD-10 codes, medical procedure code, or medication dispensing, and the cumulative incidence of AE was assessed.

Conclusion

This study provides a comprehensive and contemporary overview of patients treated by revascularization for PAD in our country in the recent years. Adjusted comparative results will allow to compare these occurrences between the different events of interest.

To our knowledge, this is the second study assessing French patients affected by PAD and treated by revascularization in a comprehensive way through French HealthCare data. The first study was carried out by EPI-PHARE between 2017 and 2019 and identified 113,766 patients having undergone angioplasty revascularization. Patients' profiles were similar to our results with a majority of men (1/3 of women) and a mean age of 71 years. Patients were also affected by comorbidities in a similar way with 80% of patients affected by a cerebral ischemic, or coronary, or peripheral artery disease, and diabetes in 40% of the population [2].

ISPOR Europe 2022 | 6-9 November 2022, Vienna, Austria

References :

[1] Fowkes FGR, Aboyans V, Fowkes FJI, McDermott MM, Sampson UKA, Criqui MH. Peripheral artery disease: epidemiology and global perspectives. Nat Rev Cardiol. 2017;14(3):156–70.

[2] Dufour E, Herlemont P, Zureik M. Stents et ballons utilisés dans l'artériopathie oblitérante des membres inférieurs (AOMI) en France entre 2017 et 2019: étude menée à partir des données du SNDS. 2021 Mar.

PMSI bases provided by ATIH, Data controller: Bayer Healthcare SAS; Processing implementation officer: Heva. Study registered under MR006 with the Health Data Hub on March 17, 2021 (Declaration of conformity No. 2220056 v 0 of October 16, 2020)

EPH15

Results

191,768 patients:
whole population

Number of patients identified per year

During the 4-year study period, a total of 191,768 patients underwent revascularization for PAD and were included in the study. From 55,151 to 58,493 patients underwent revascularization for PAD each year. Patients had a mean age of 72 (±12.2) years at index date and were mainly men (69.2%).

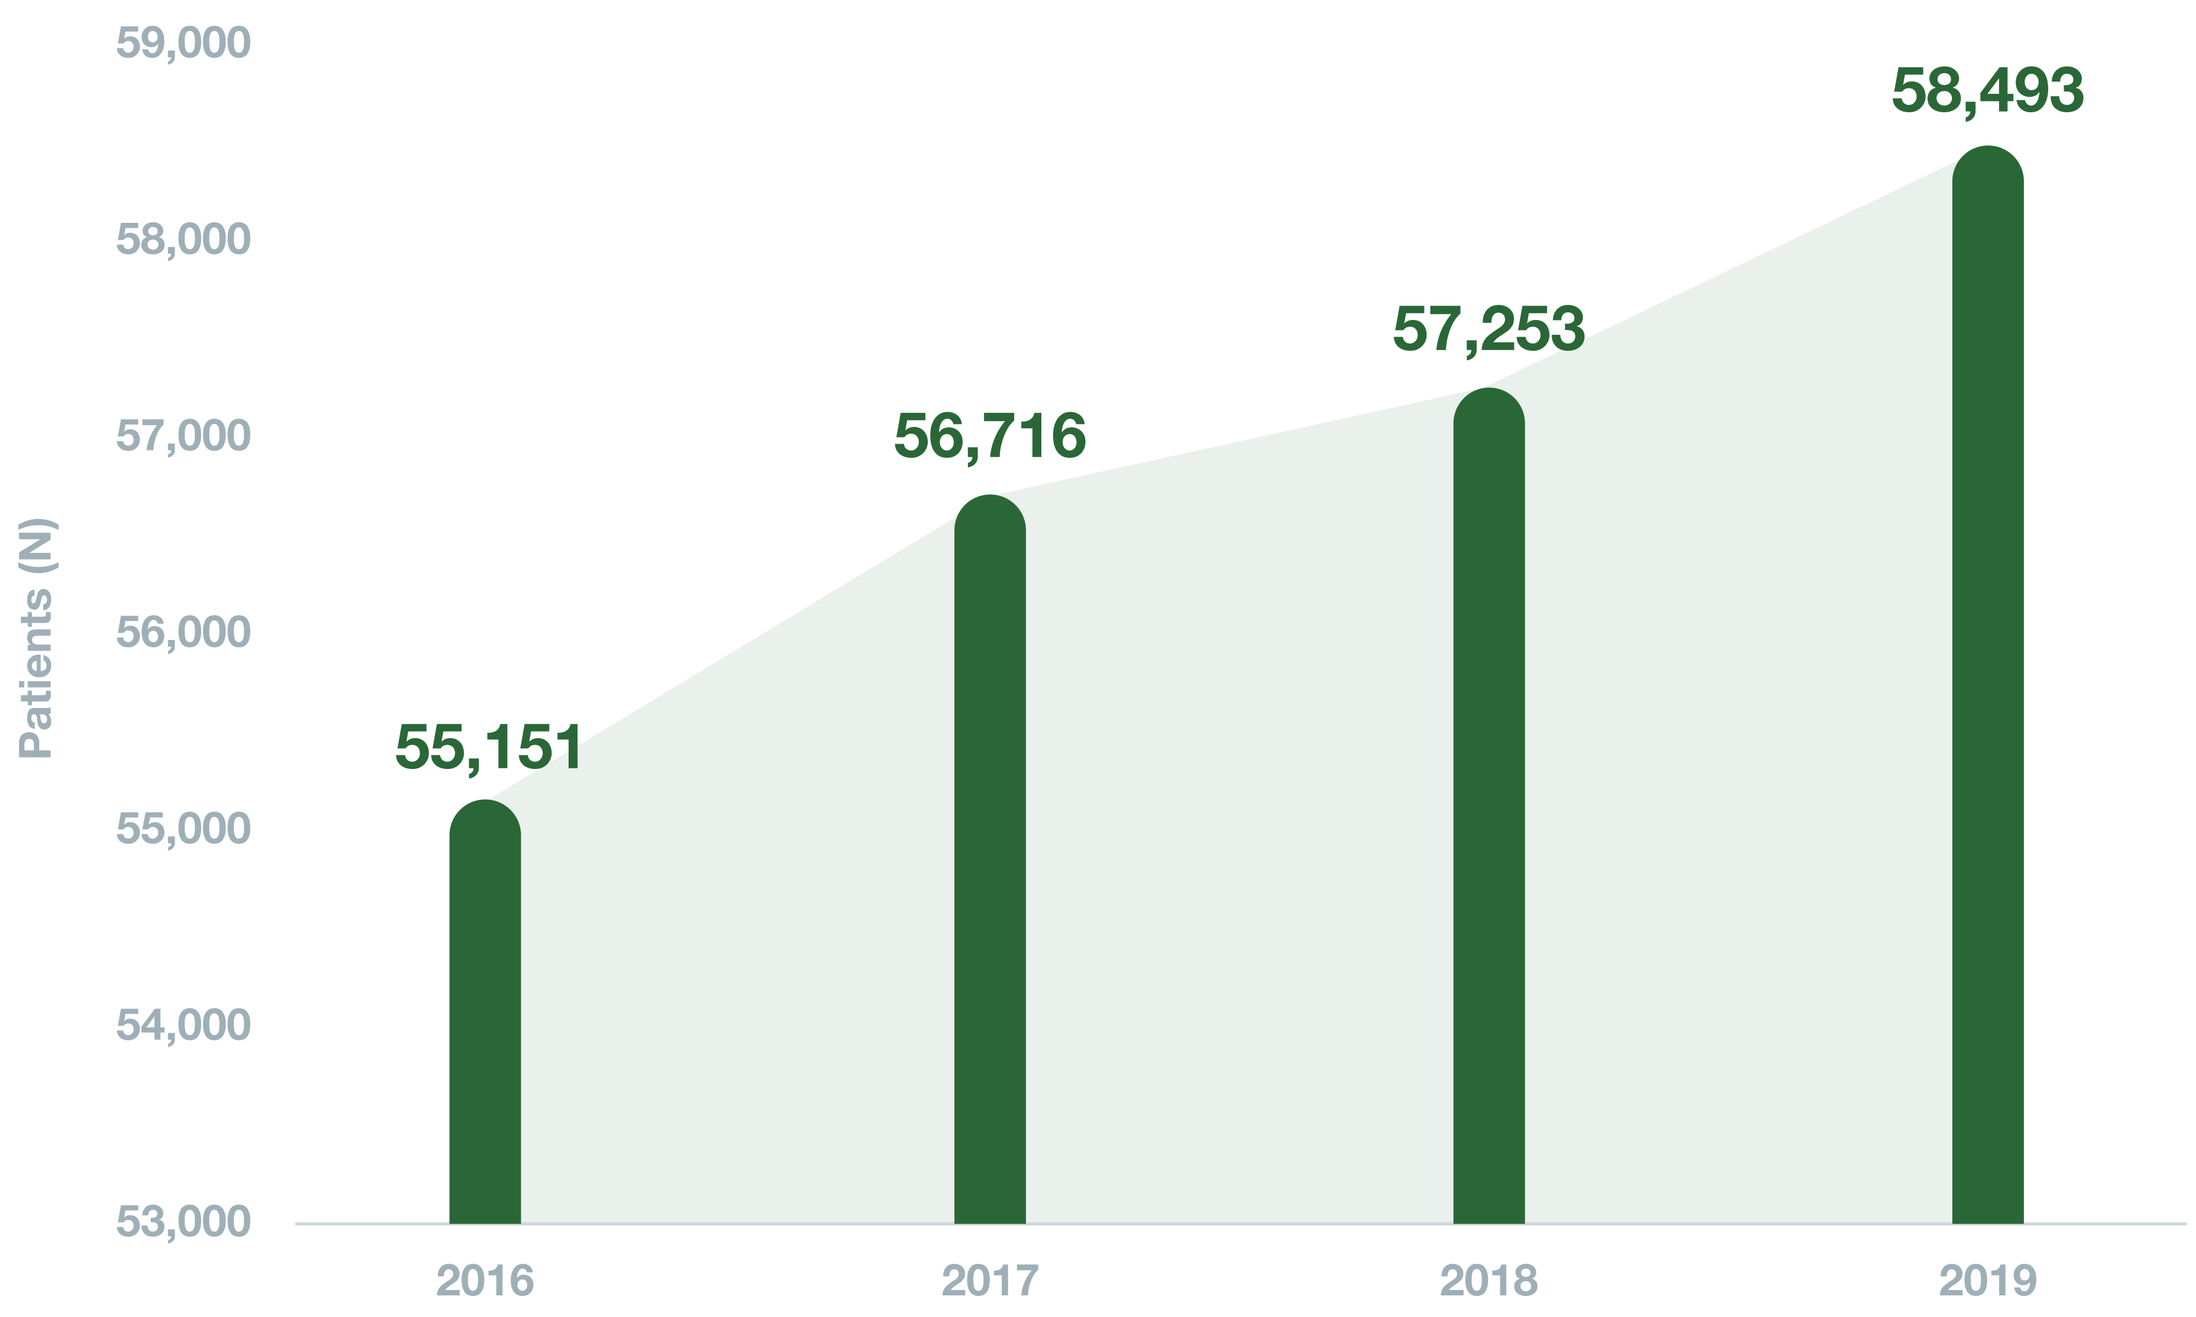
59,000
58,000
57,000
56,000
55,000
54,000
53,000
Patients (N)
55,151
56,716
57,253
58,493
2016
2017
2018
2019

Patients undergoing a procedure during the year of interest independently of revascularization history

Comorbidities distribution and medical history

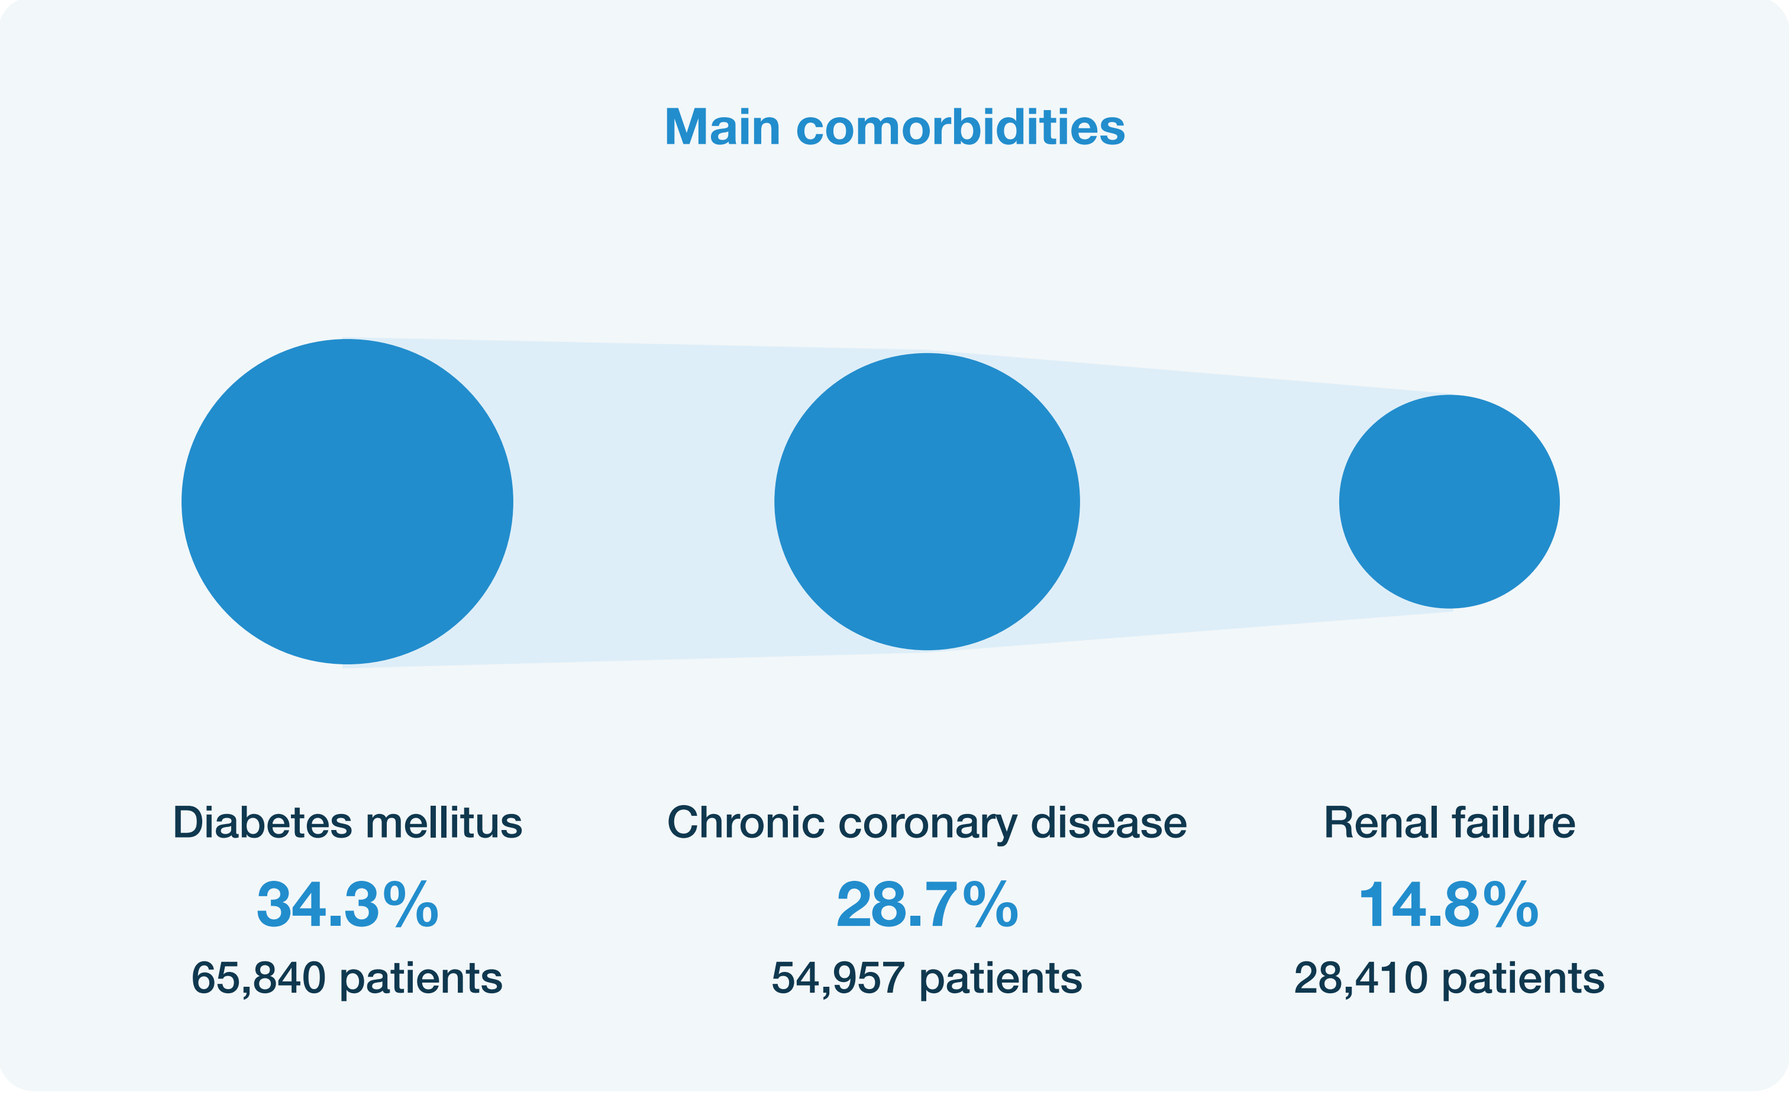
Main comorbidities
Diabetes mellitus
34.3%
65,840 patients
Chronic coronary disease
28.7%
54,957 patients
Renal failure
14.8%
28,410 patients

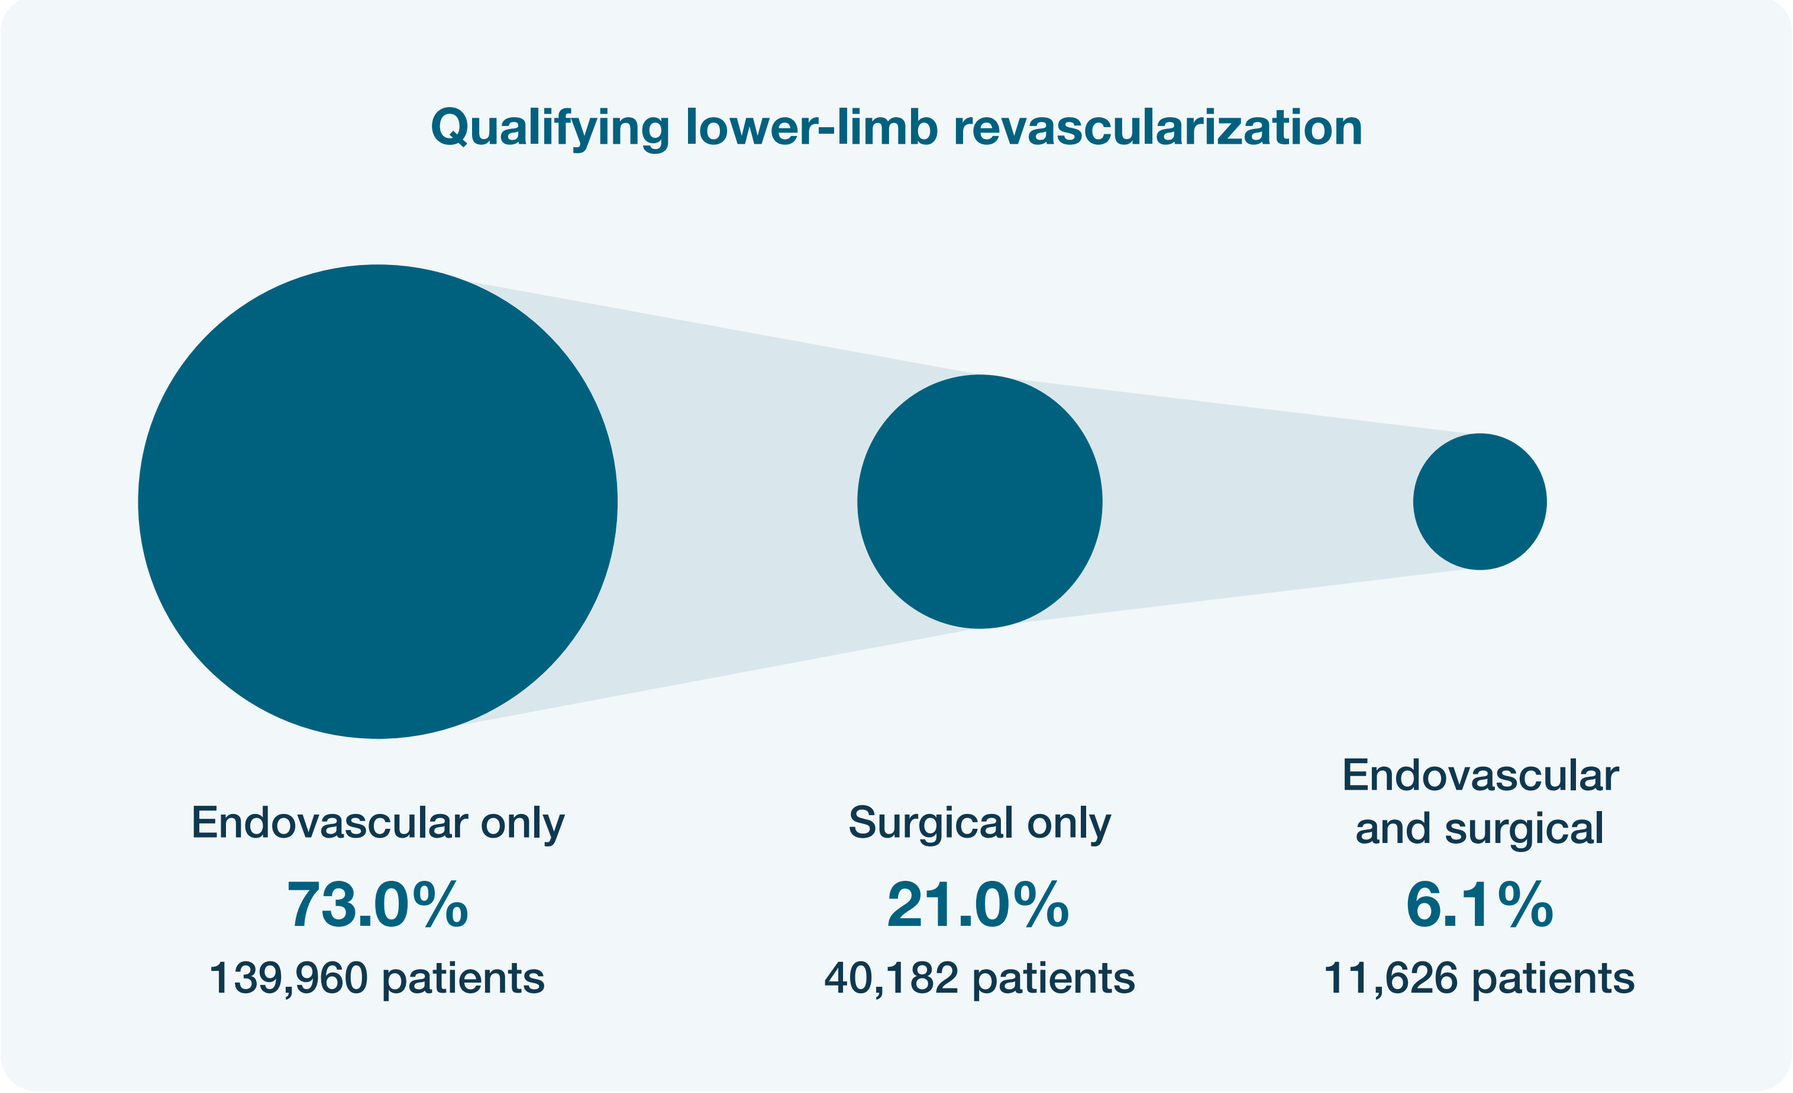
Qualifying lower-limb revascularization
Endovascular only
73.0%
139,960 patients
Surgical only
21.0%
40,182 patients
Endovascular and surgical
6.1%
11,626 patients

The three most frequent comorbidities were diabetes mellitus (34.3%), chronic coronary disease (28.7%), and renal failure (14.8%).

148,132 patients:
patients with at least a year of follow-up

Cumulative incidence of the first atherothrombotic event

(secondary revascularization excluded)

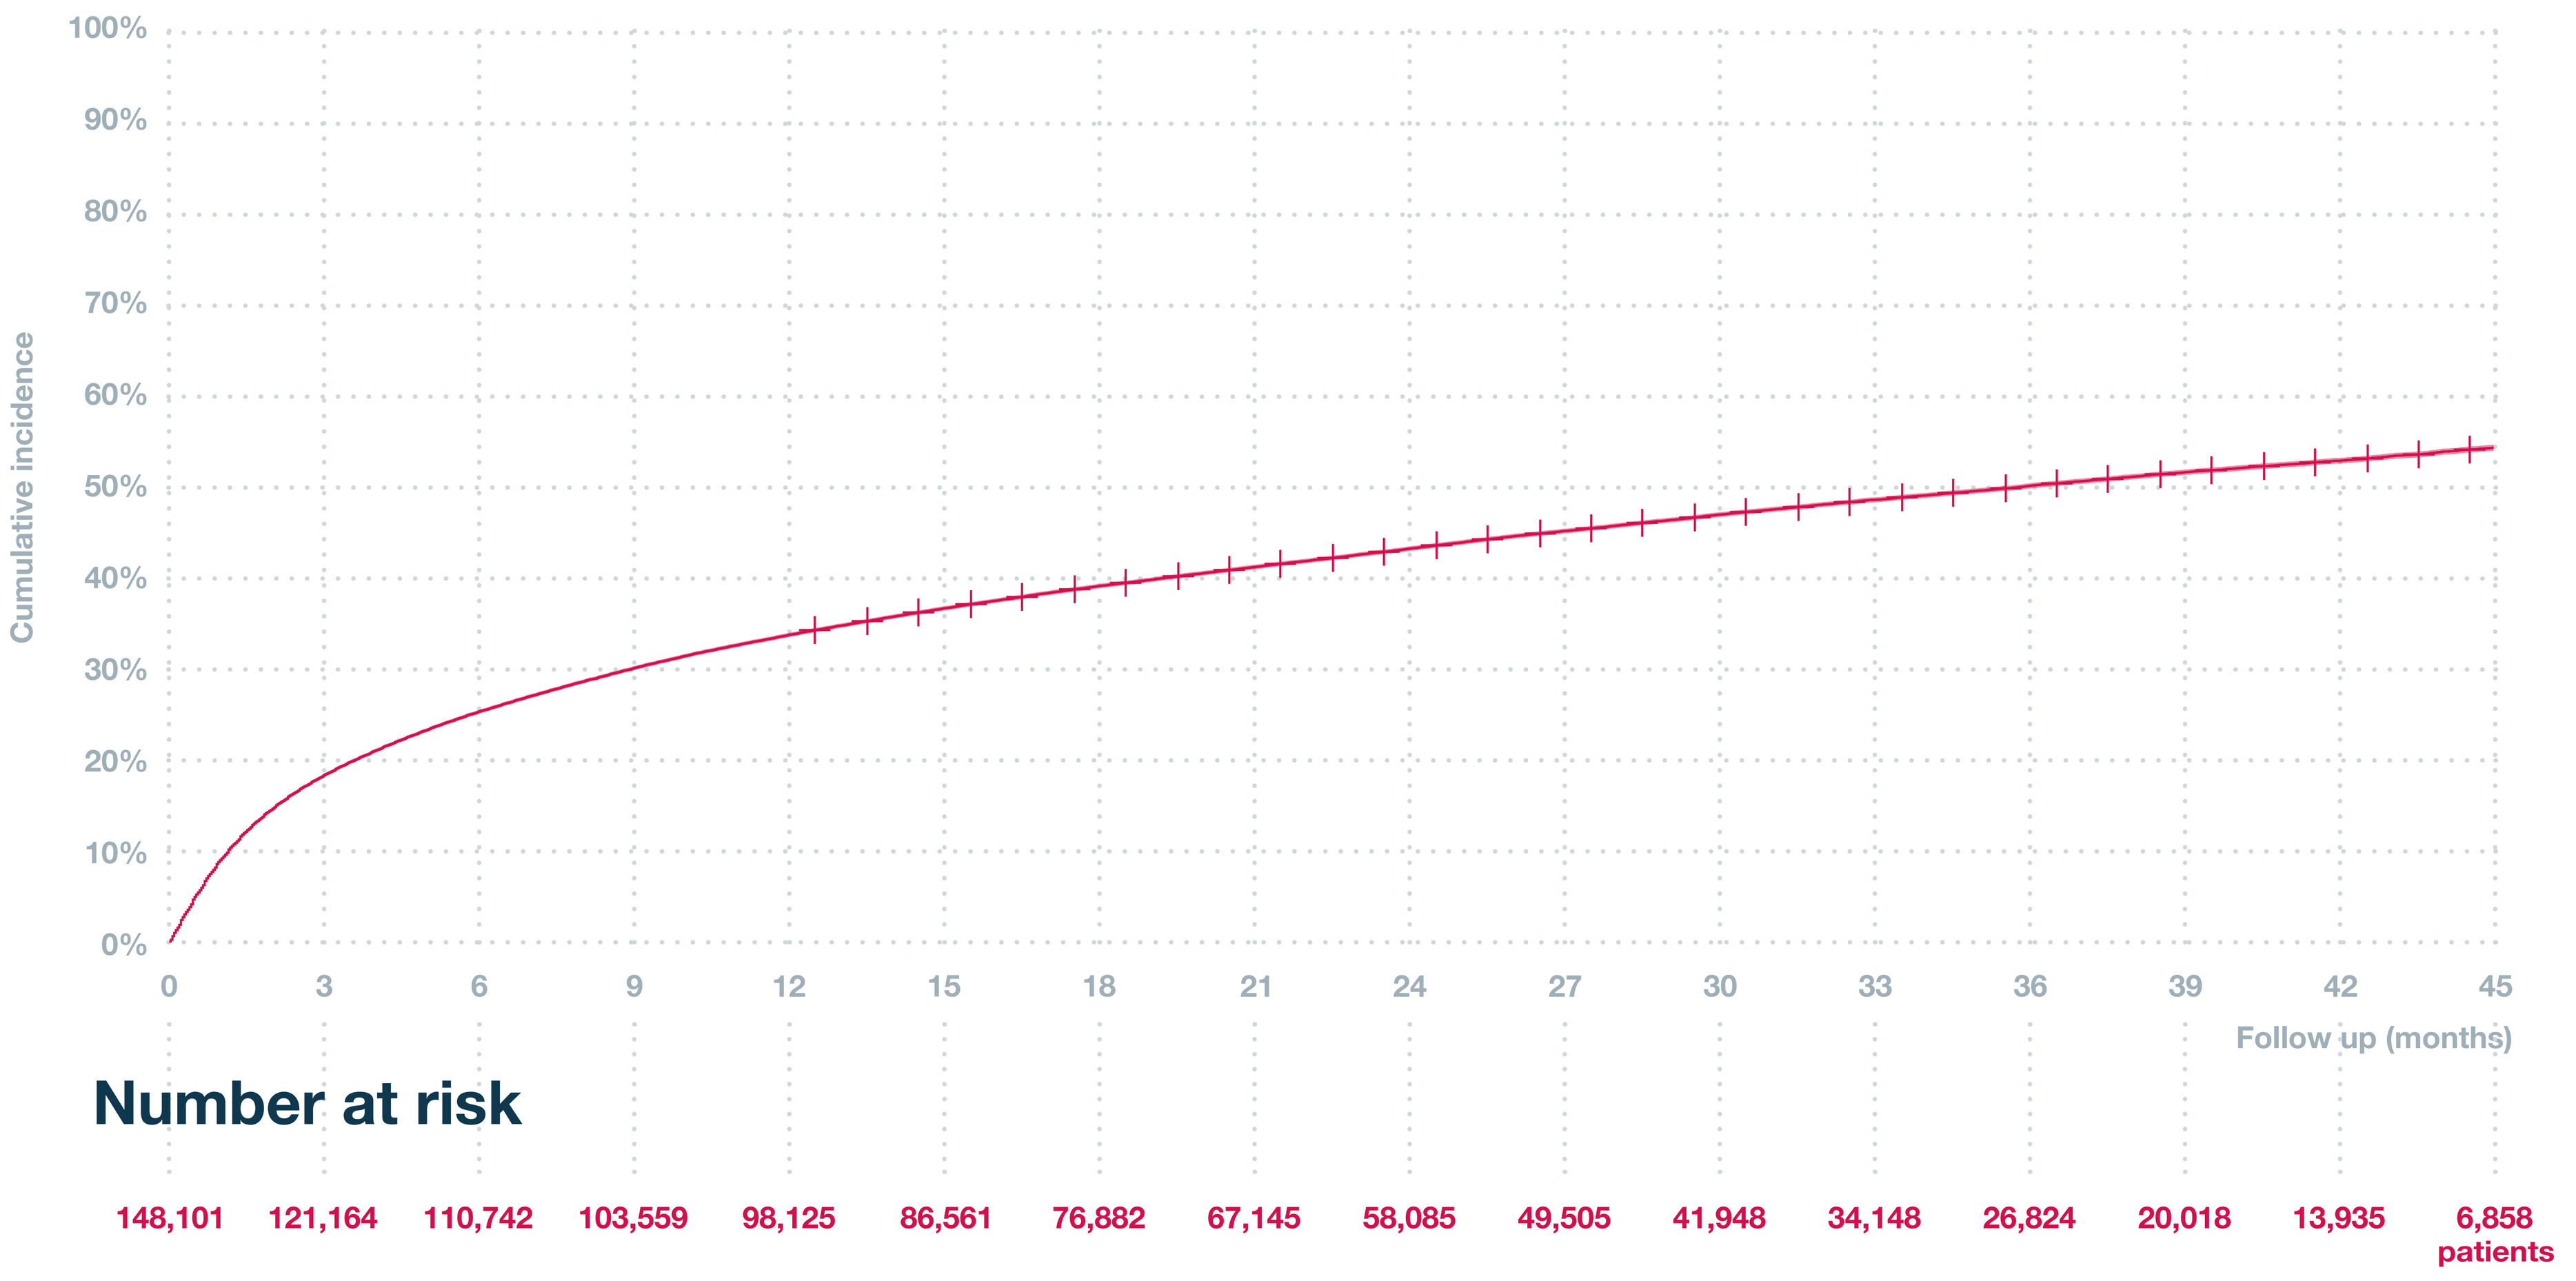
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Cumulative incidence
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Follow up (months)
Number at risk
148,101 121,164 110,742 103,559 98,125 86,561 76,882 67,145 58,085 49,505 41,948 34,148 26,824 20,018 13,935 6,858 patients

148,132 patients had at least one year of follow-up. Cumulative incidence of the first AE at 12, 24 and 36 months were 33.8%, 43.2% and 50.1%.

Atherothrombotic events

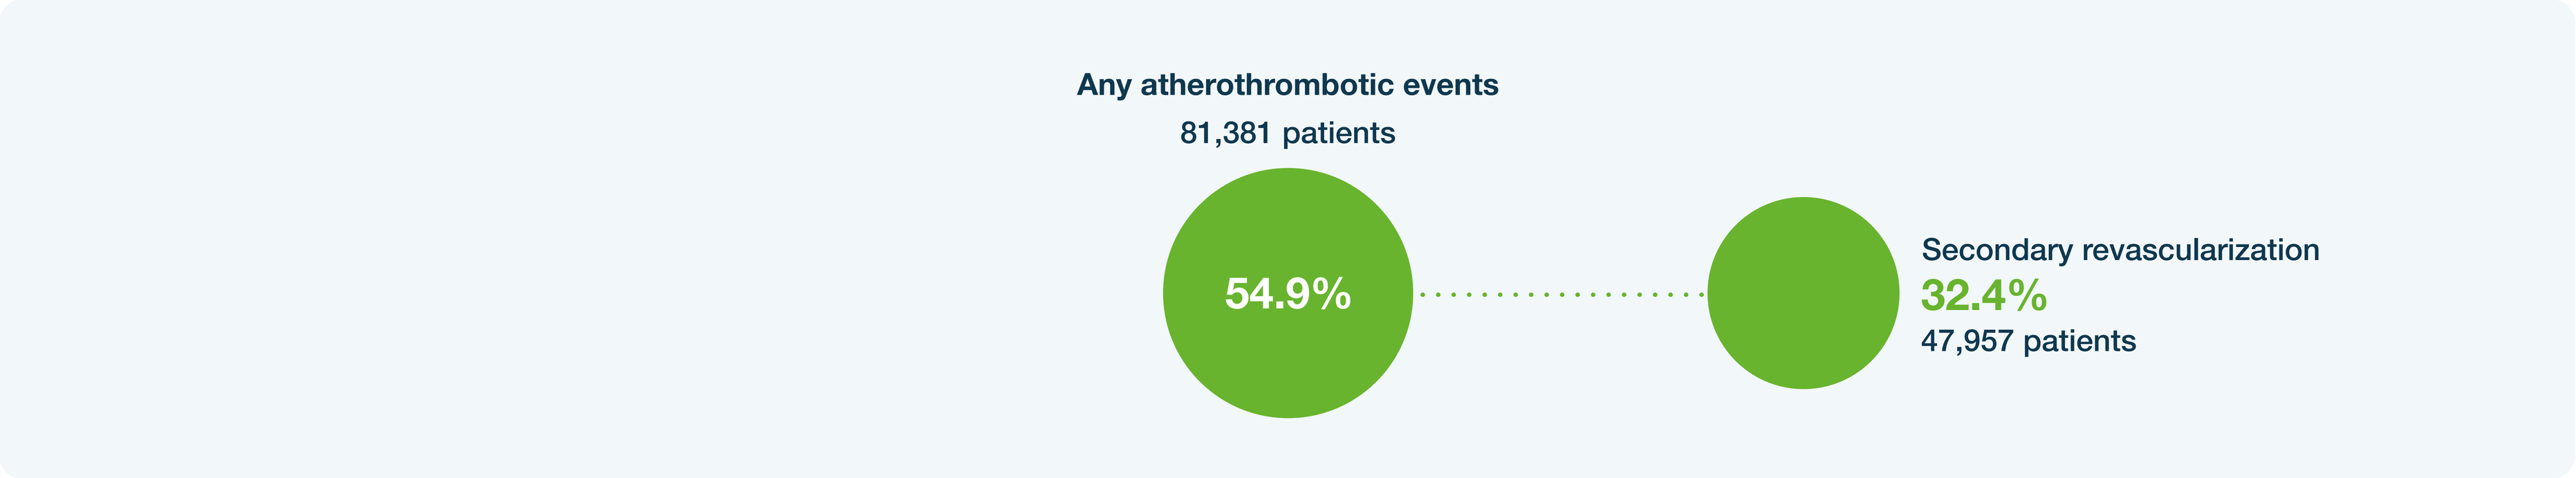
Any atherothrombotic events
81,381 patients
54.9%
Secondary revascularization
32.4%
47,957 patients

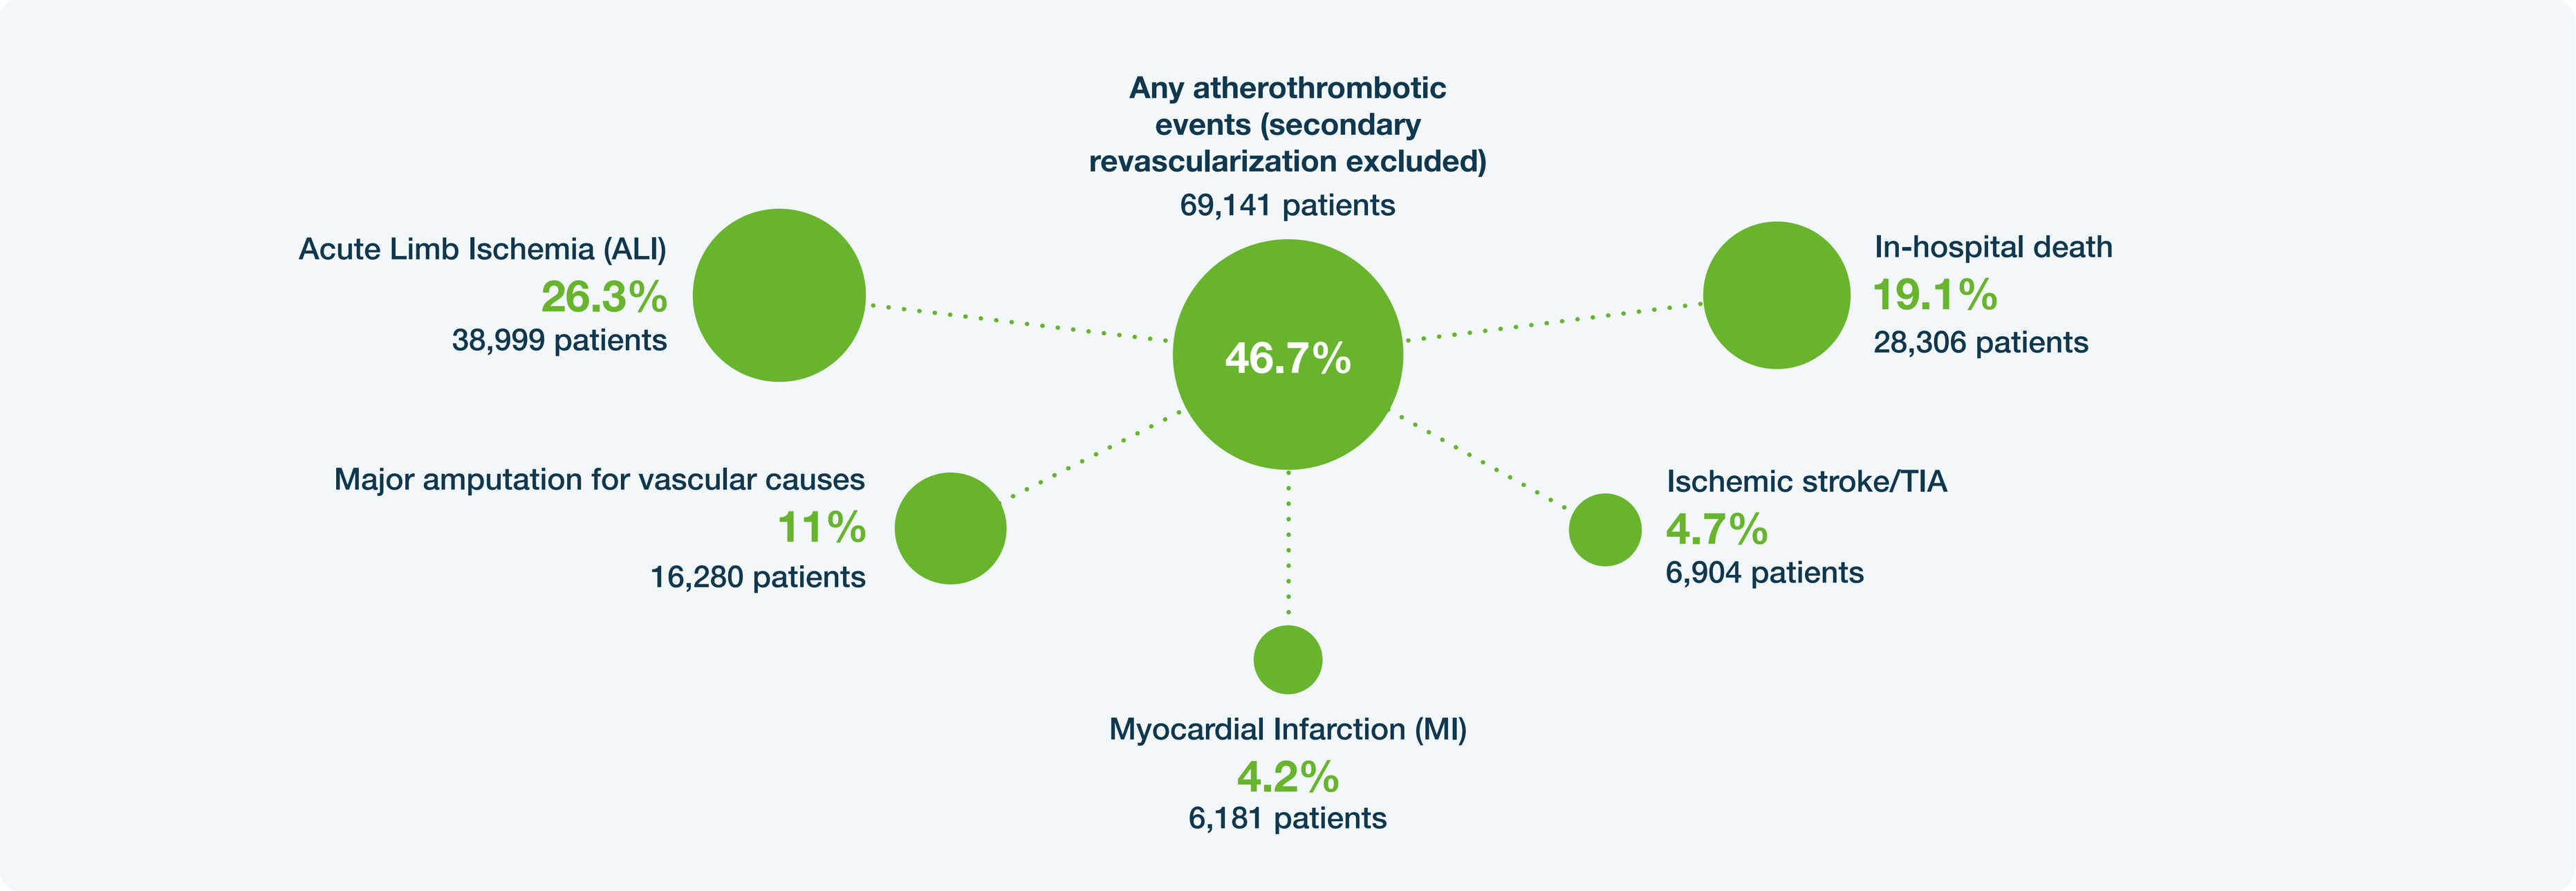
Any atherothrombotic events (secondary revascularization excluded)
69,141 patients
46.7%
Acute Limb Ischemia (ALI)
26.3%
38,999 patients
In-hospital death
19.1%
28,306 patients
Major amputation for vascular causes
11%
16,280 patients
Ischemic stroke/TIA
4.7%
6,904 patients
Myocardial Infarction (MI)
4.2%
6,181 patients

The three most frequent events that occurred were ALI (26.3%), all-cause in-hospital death (19.1%), and major amputation for vascular causes (11.0%). Secondary revascularization occurred in 32.4% of patients.